Poster #P335

Rezafungin Safety and Pharmacokinetics in Subjects with Moderate or Severe Hepatic Impairment

Shawn Flanagan[1], Voon Ong[1], Taylor Sandison[1], Rebeca Melara[2], Thomas Marbury[3], Alena Jandourek[1] and Jade Huguet[2]

[1]Cidara Therapeutics Inc.; [2]Altasciences; [3]Orlando Clinical Research Center

Taylor Sandison
Cidara Therapeutics, Inc.
6310 Nancy Ridge Drive, Suite 101
San Diego, CA, USA
tsandison@cidara.com

INTRODUCTION

Rezafungin is a novel echinocandin antifungal being developed for treatment of candidemia/invasive candidiasis, and for prevention of invasive fungal disease caused by *Candida* and *Aspergillus* spp. and *Pneumocystis jirovecii* in immunosuppressed patients following BMT.

This study was designed to assess the safety and pharmacokinetics (PK) of rezafungin in subjects with normal hepatic function and subjects with moderate or severe hepatic impairment, as rezafungin clinical trials have been limited to enrolling patients with moderate hepatic impairment due to the comparator caspofungin's product label.

METHODS

The safety, tolerability, and PK of rezafungin in subjects with moderate (Group 1a) and severe (Group 2a) hepatic impairment (Child-Pugh class B and C) and healthy subjects (Groups 1b, and 2b) was investigated in this open-label, single-dose study.

Each hepatic impairment group was matched to a separate healthy subject group (n=8 each, 32 total) by age, sex, and body mass index (BMI) and received a single 400-mg intravenous 1-hour infusion of RZF.

Plasma PK sampling was performed at various time points through 2 weeks post-dose. Rezafungin PK parameters were derived using non-compartmental analysis. Safety and tolerability were assessed throughout the study.

RESULTS

- Subjects with moderate or severe hepatic impairment were well matched to their respective heathy subject groups (Table 1).
- There were 9 adverse events (AEs) in 7 subjects:
 - 3 occurred in the severe hepatic impairment group (bronchitis, worsening hepatic encephalopathy, hyponatremia), all of moderate severity and unrelated to RZF
 - 5 occurred in the moderate hepatic impairment group (nausea, headache, infusion site infiltration, body aches, productive cough), all of mild severity and only 1 AE was considered related to RZF (headache)
 - 1 AE occurred in the healthy group (infusion site infiltration), of mild severity and unrelated to RZF
- All AEs resolved or were resolving at the end of the study.
- Mean RZF exposure in subjects with moderate or severe hepatic impairment was up to ~30% lower than that in healthy subjects, with considerable overlap between individuals (Figure 1A, 1B). These differences were not considered to be clinically relevant. Mean half-life values were similar between groups and ranged from 110 to 124 hours.

Table 1. Subject demographics and characteristics in hepatic impairment and matched healthy groups

Parameter	Stage 1		Stage 2		Overall (N = 34)
	Moderate hepatic impairment (N = 8)	Normal hepatic function (N = 8)	Severe hepatic impairment (N = 8)	Normal hepatic function (N = 8)	
Age, years					
Mean (SD)	57.0 (4.47)	56.9 (4.94)	58.0 (9.23)	56.6 (4.81)	57.1 (5.90)
Median (Min, Max)	56.5 (50, 63)	56.0 (50, 65)	61.5 (41, 68)	57.5 (50, 61)	57.0 (41, 68)
Sex, n (%)					
Male	4 (50.0)	4 (50.0)	6 (75.0)	6 (75.0)	20 (62.5)
Female	4 (50.0)	4 (50.0)	2 (25.0)	2 (25.0)	12 (37.5)
Ethnicity, n (%)					
Hispanic/Latino	0	2 (25.0)	2 (25.0)	1 (12.5)	5 (15.6)
Not Hispanic/Not Latino	8 (100.0)	6 (75.0)	6 (75.0)	7 (87.5)	27 (84.4)
Race, n (%)					
White	7 (87.5)	5 (62.5)	7 (87.5)	4 (50.0)	23 (71.9)
Black or African American	1 (12.5)	2 (25.0)	0	4 (50.0)	7 (21.9)
Asian	0	1 (12.5)	0	0	1 (3.1)
Other	0	0	1 (12.5)	0	1 (3.1)
Weight,kg					
Mean (SD)	98.09 (21.348)	88.55 (7.234)	87.55 (11.600)	85.11 (9.386)	89.82 (13.788)
Median (Min, Max)	91.25 (72.8, 136.7)	89.65 (76.8, 99.1)	87.65 (74.3, 106.3)	84.00 (71.8, 98.0)	88.35 (71.8, 136.7)
Height, cm					
Mean (SD)	174.09 (12.386)	168.76 (8.003)	171.95 (7.170)	169.40 (4.912)	171.05 (8.415)
Median (Min, Max)	172.75 (158.6, 195.5)	169.20 (157.5, 181.0)	173.50 (161.5, 181.5)	168.40 (163.0, 178.0)	169.95 (157.5, 195.5)
Body Mass Index, kg/m^2					
Mean (SD)	32.04 (3.449)	31.14 (2.361)	29.64 (3.681)	29.66 (3.007)	30.62 (3.181)
Median (Min, Max)	32.35 (26.7, 35.8)	30.85 (28.5, 35.2)	30.55 (23.5, 34.4)	29.35 (25.3, 33.6)	30.80 (23.5, 35.8)

Figure 1. Individual and Geometric Mean (95% Confidence Interval) Rezafungin (A) AUC_{inf} and (B) C_{max}

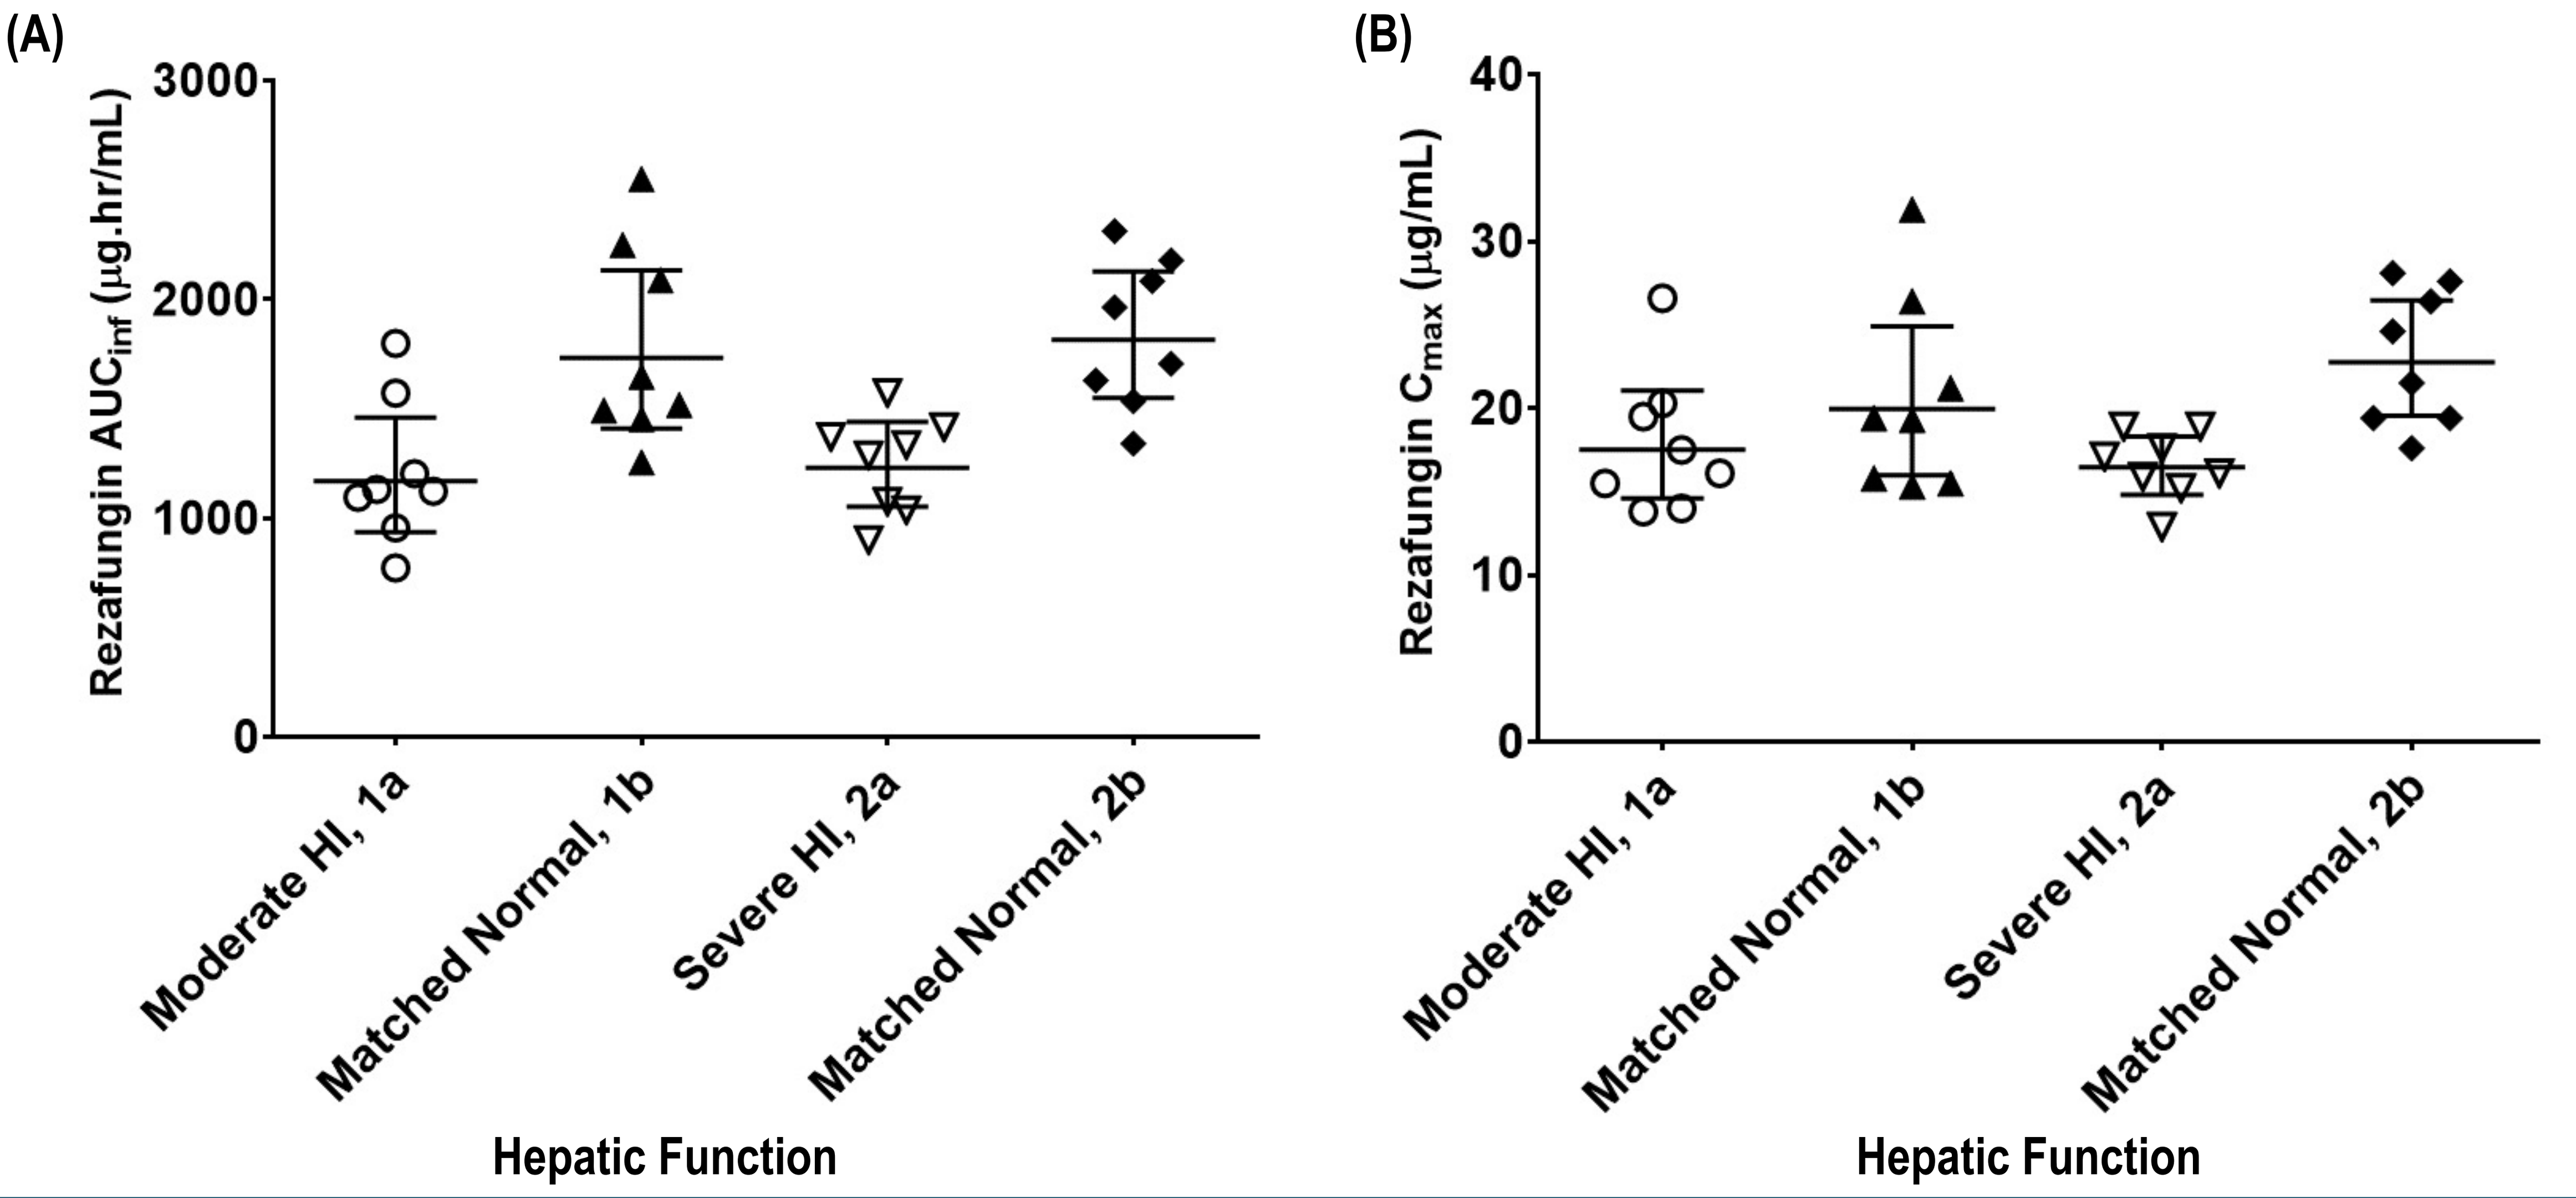
(A)
Rezafungin AUCinf (µg.hr/mL)
3000
2000
1000
0
Moderate HI, 1a
Matched Normal, 1b
Severe HI, 2a
Matched Normal, 2b
Hepatic Function
(B)
Rezafungin Cmax (µg/mL)
40
30
20
10
0
Moderate HI, 1a
Matched Normal, 1b
Severe HI, 2a
Matched Normal, 2b
Hepatic Function

CONCLUSIONS

- Rezafungin exposure was modestly reduced on average in subjects with moderate or severe hepatic impairment relative to matched healthy subjects, with considerable overlap between individuals.
- There were no significant safety findings associated with rezafungin in any group.
- More AEs were experienced by subjects with moderate or severe hepatic impairment than healthy subjects, as expected given their underlying liver disease.
- These results indicate that rezafungin can be administered to subjects with all levels of hepatic impairment without dose adjustment.

DISCLOSURES / ACKNOWLEDGEMENTS

This study was funded by Cidara Therapeutics and Mundipharma. Editorial assistance was provided by T. Chung (Cidara Therapeutics).