Discovery of CBO-212, a first-in-class Drug Fc-Conjugate (DFC), targeting CD73 in Cancer

Poster 270

(45P)

James Levin[1], Simon Döhrmann[1], Nicholas Dedeic, Amanda Almaguer, Doug Zuill, Elizabeth Abelovski, Rajvir Grewal, Joanne Fortier, Qiping Zhao, Maria Hernandez, Karin Amundson, Madison Moniz, Hongyuan Chen, Travis Haussener, Dhanya Panickar, Thanh Lam, Thomas P. Brady, Allen Borchardt, Jason N. Cole, and Leslie W. Tari.

[1]Presenting authors; Cidara Therapeutics, 6310 Nancy Ridge Drive, San Diego, CA, 92121 USA

James Levin (Jlevin@Cidara.com)

Simon Döhrmann (Sdoehrmann@Cidara.com)

INTRODUCTION

Cidara Therapeutics is developing a new generation of immune modulating anti-tumor agents to treat solid tumors. CD73 contributes to immune evasion in solid tumors by producing immune-suppressive adenosine in the tumor microenvironment (TME). Herein, we describe CBO-212, a first-in-class CD73 targeting drug-Fc conjugate (DFC), comprising a multi-valent conjugate of a novel small molecule CD73 inhibitor to a proprietary immune-silent hIgG1 Fc (**Figure 1**). CBO-212 combines the strengths of small molecule inhibitors and monoclonal antibodies (mAb) targeting CD73 currently in clinical development, with potential best-in-class activity.

Figure 1. *Schematic difference between a mAb and DFC*

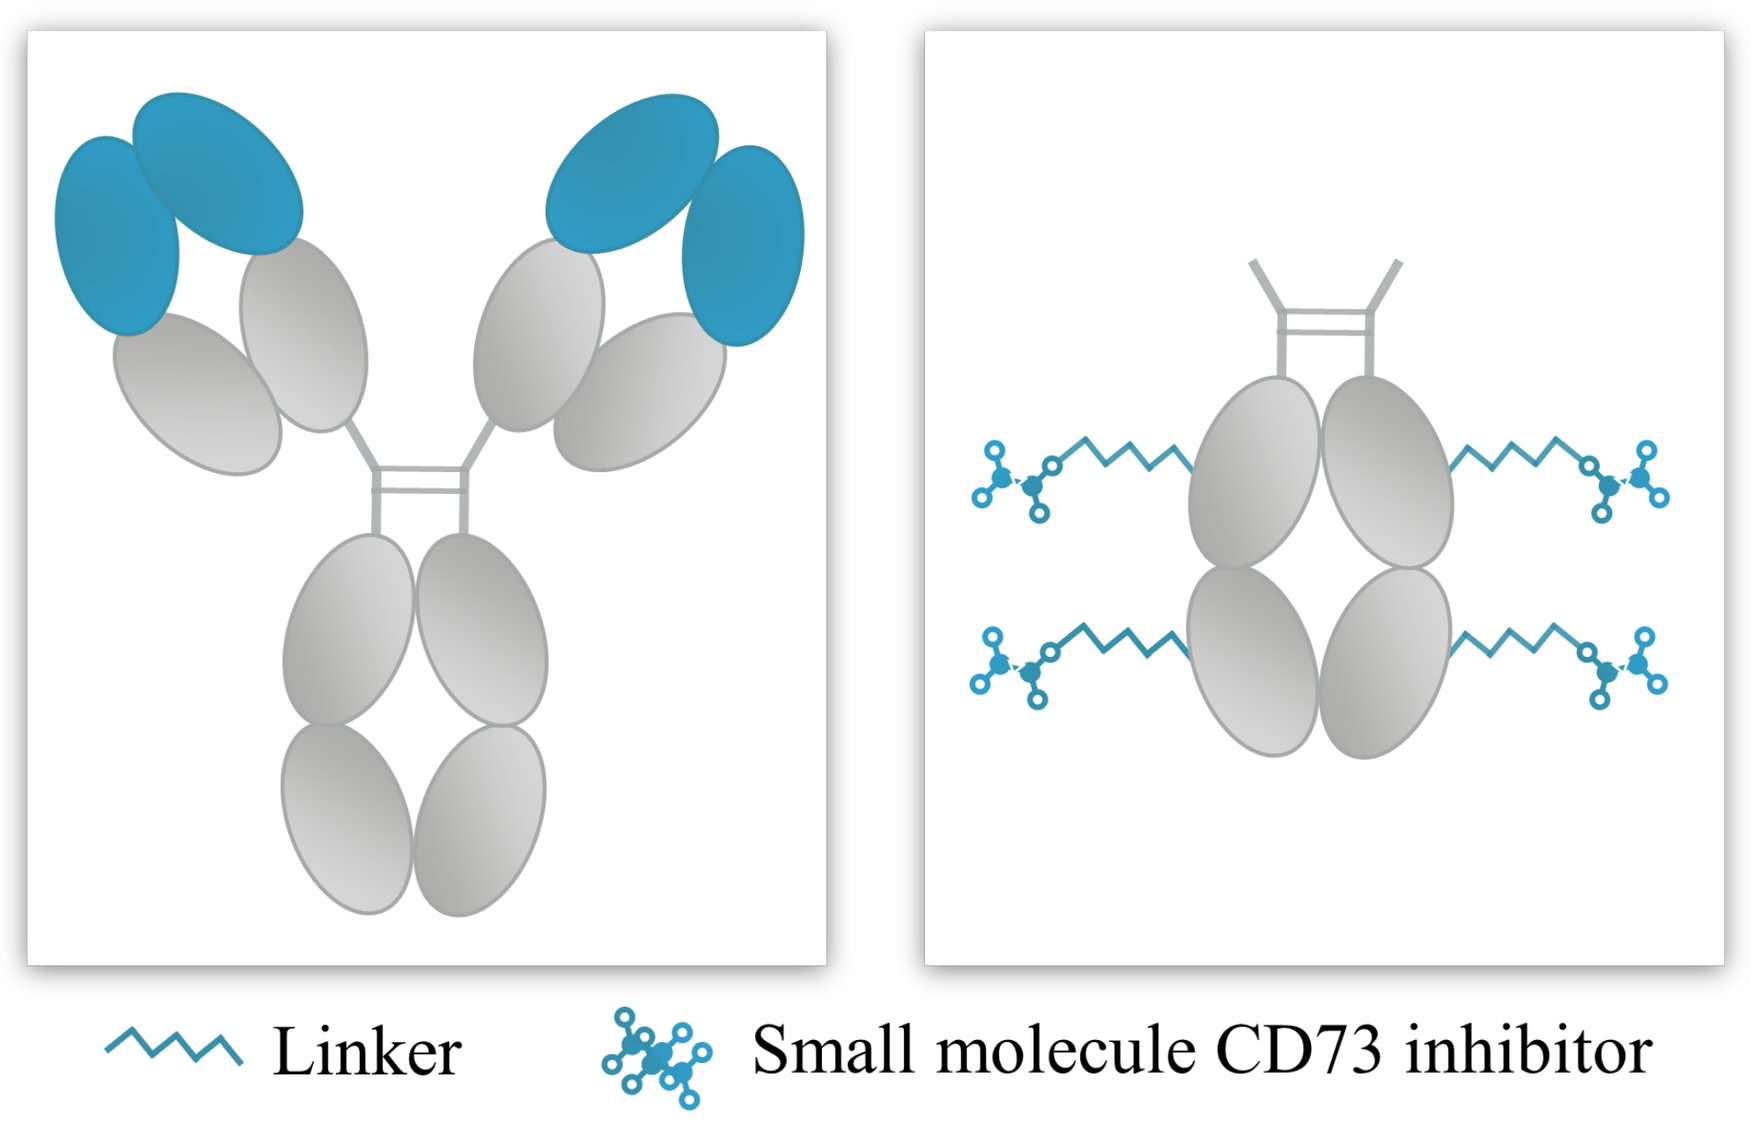
mAb
DFC
Linker
Small molecule CD73 inhibitor

METHODS

Inhibition of CD73 was evaluated using cell-free and cell-based assays. Functional activity was measured in PBMC rescue assays in the presence of AMP by flow cytometry. CD73 internalization was determined in MDA-MB-231 cells.

Pharmacokinetic (PK) studies were conducted in BALB/c mice. Plasma concentrations and conjugate stability were measured using sandwich ELISA with human CD73 capture and anti-human IgG detection. Efficacy of CBO-212 was evaluated in a syngeneic mouse model with established CT26 tumors and utilized a single IP administration of CBO-212 at 20 mg/kg.

RESULTS

Figure 2. *Soluble CD73 inhibition assay (IC_{50} in nM)*

Test article	IC_{50} [nM]
CBO-212	9.11
AB680	1.93
OP-5244	1.28
SHR170008	4.02
Oleclumab	n.c.
Mupadolimab	>100

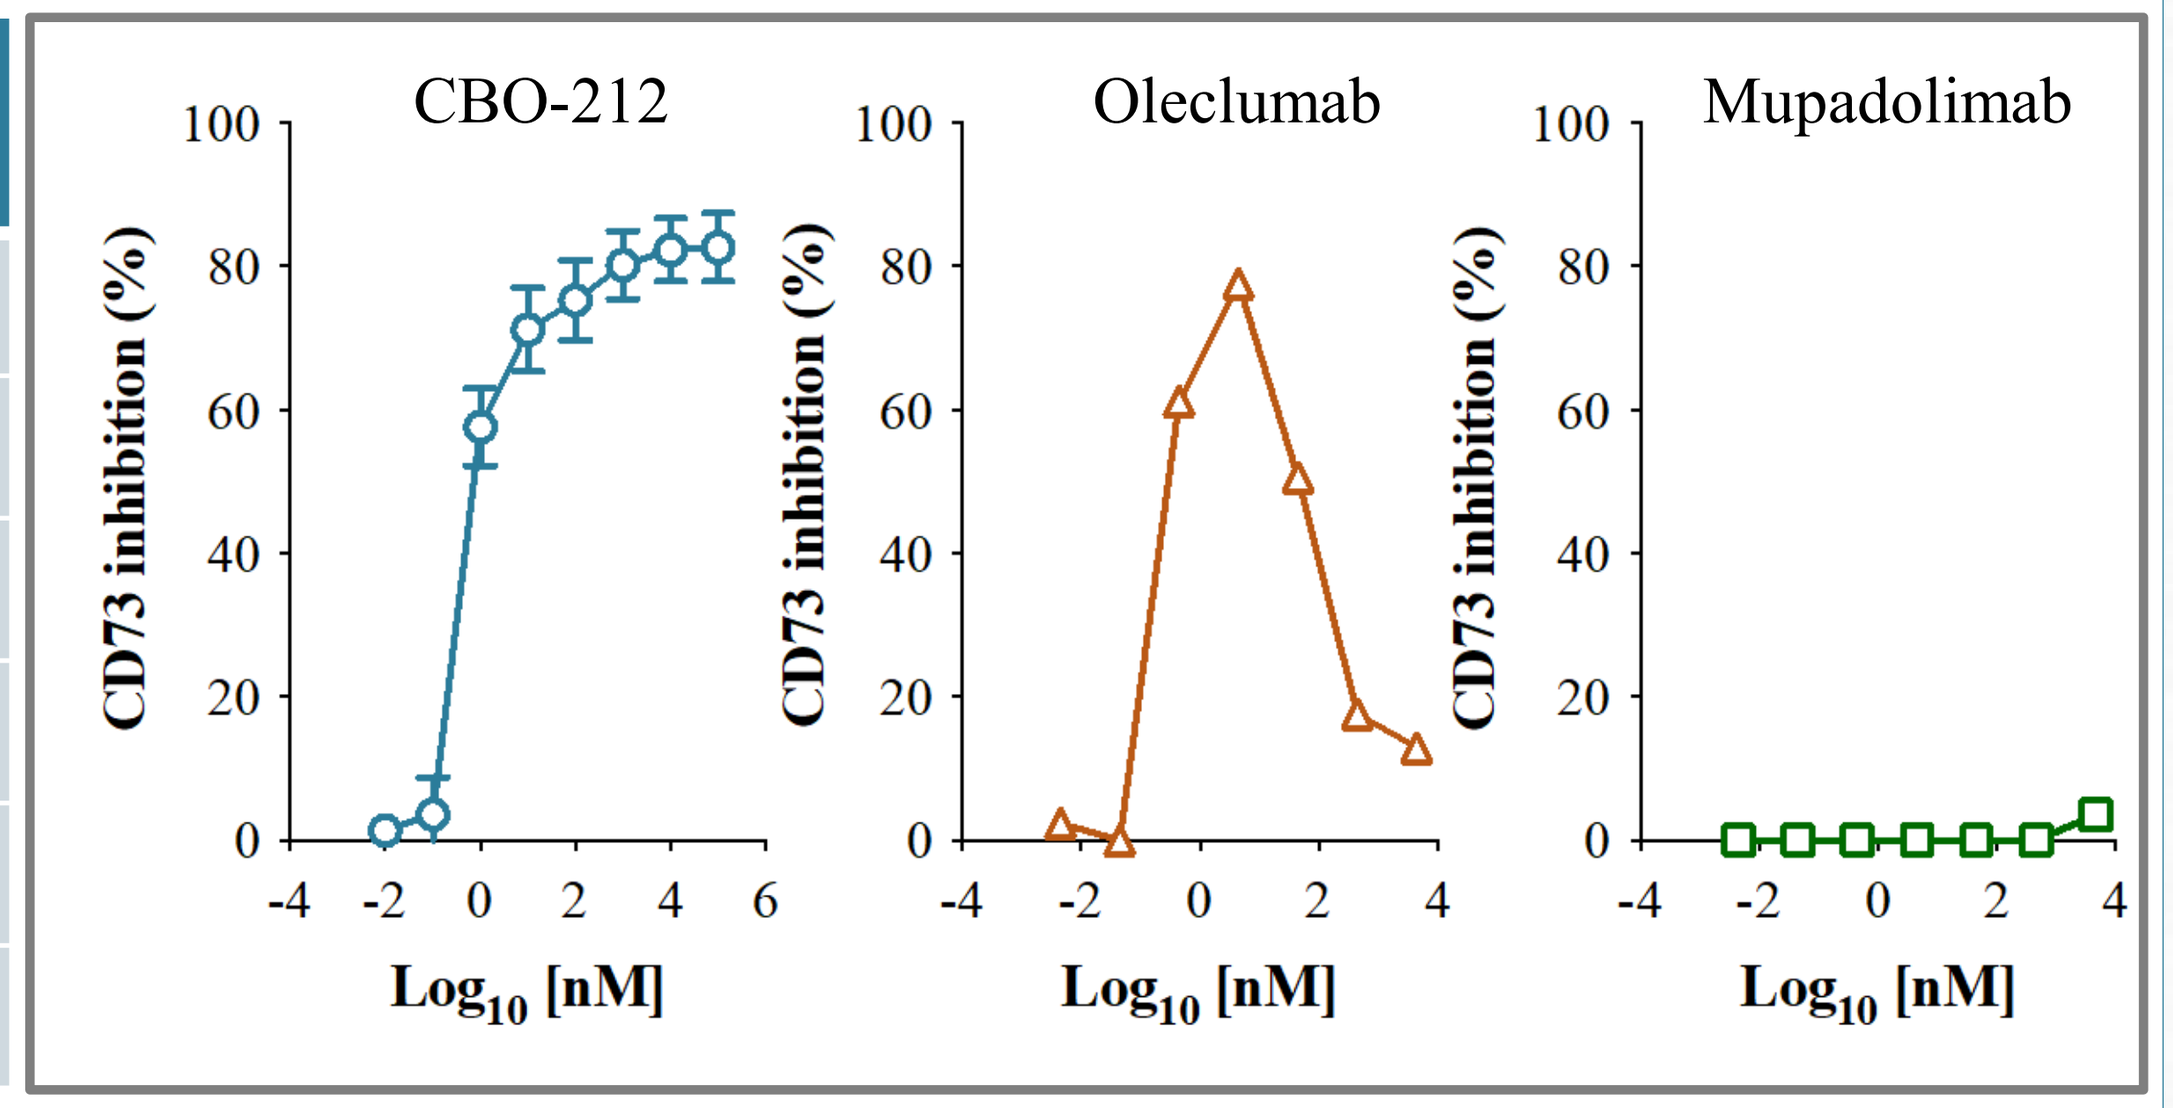
CBO-212
Oleclumab
Mupadolimab
CD73 inhibition (%)
Log10 [nM]

RESULTS (cont.)

CD73 is an ecto-nucleotidase found in cell-anchored and soluble forms. Both forms contribute to formation of immune-suppressive adenosine in the TME. We evaluated CBO-212 inhibition of catalytic activity against both forms. CBO-212 was a potent inhibitor of soluble CD73 (**Figure 2**). Unlike the mAb comparators, CBO-212 was a full inhibitor of soluble CD73 catalytic activity.

Table 1. *Cell-based CD73 inhibition assay (IC_{50} in nM)*

Test article	Human MDA-MB-231	Human PBMCs median (range, n=3)	Murine 4T1
CBO-212	0.55	0.8 (0.46 – 2.07)	6.21
AB680	0.34	0.05 (0.04 – 0.09)	6.79
OP-5244	0.31	0.07 (0.04 – 0.08)	1.67
SHR170008	4.87	1.45 (1.14 – 1.49)	8.20
Oleclumab	0.11	7.26 (3.12 – 16.6)	>100
Mupadolimab	1.16	9.27 (2.4 – 9.78)	>100

Multivalent inhibitor presentation with CBO-212 appears to enhance its activity against cell-anchored versus soluble CD73. Against CD73 expressed on a human breast cancer line (MDA-MB-231), CBO-212 activity is similar to small molecule and mAb inhibitors (**Table 1**). Against CD73 expressed on human PBMCs, CBO-212 demonstrated enhanced potency versus comparator mAbs. Against a mouse breast cancer line that expresses CD73 at high levels (4T1), CBO-212 was significantly more potent than both mAbs (mupadolimab does not bind to murine CD73) and displayed similar activity to the small molecule inhibitors. Collectively, CBO-212 showed potent inhibition of soluble and cell-anchored CD73.

Figure 3. *PBMC rescue assay of AMP suppressed cells (EC_{50} in nM)*

Test article	CD25	Granzyme B
CBO-212	11.6	1.2
AB680	8.1	1.4
OP-5244	5.9	1.4
SHR170008	597.7	212.2
Oleclumab	>1,000	>1,000
Mupadolimab	932.3	63.1

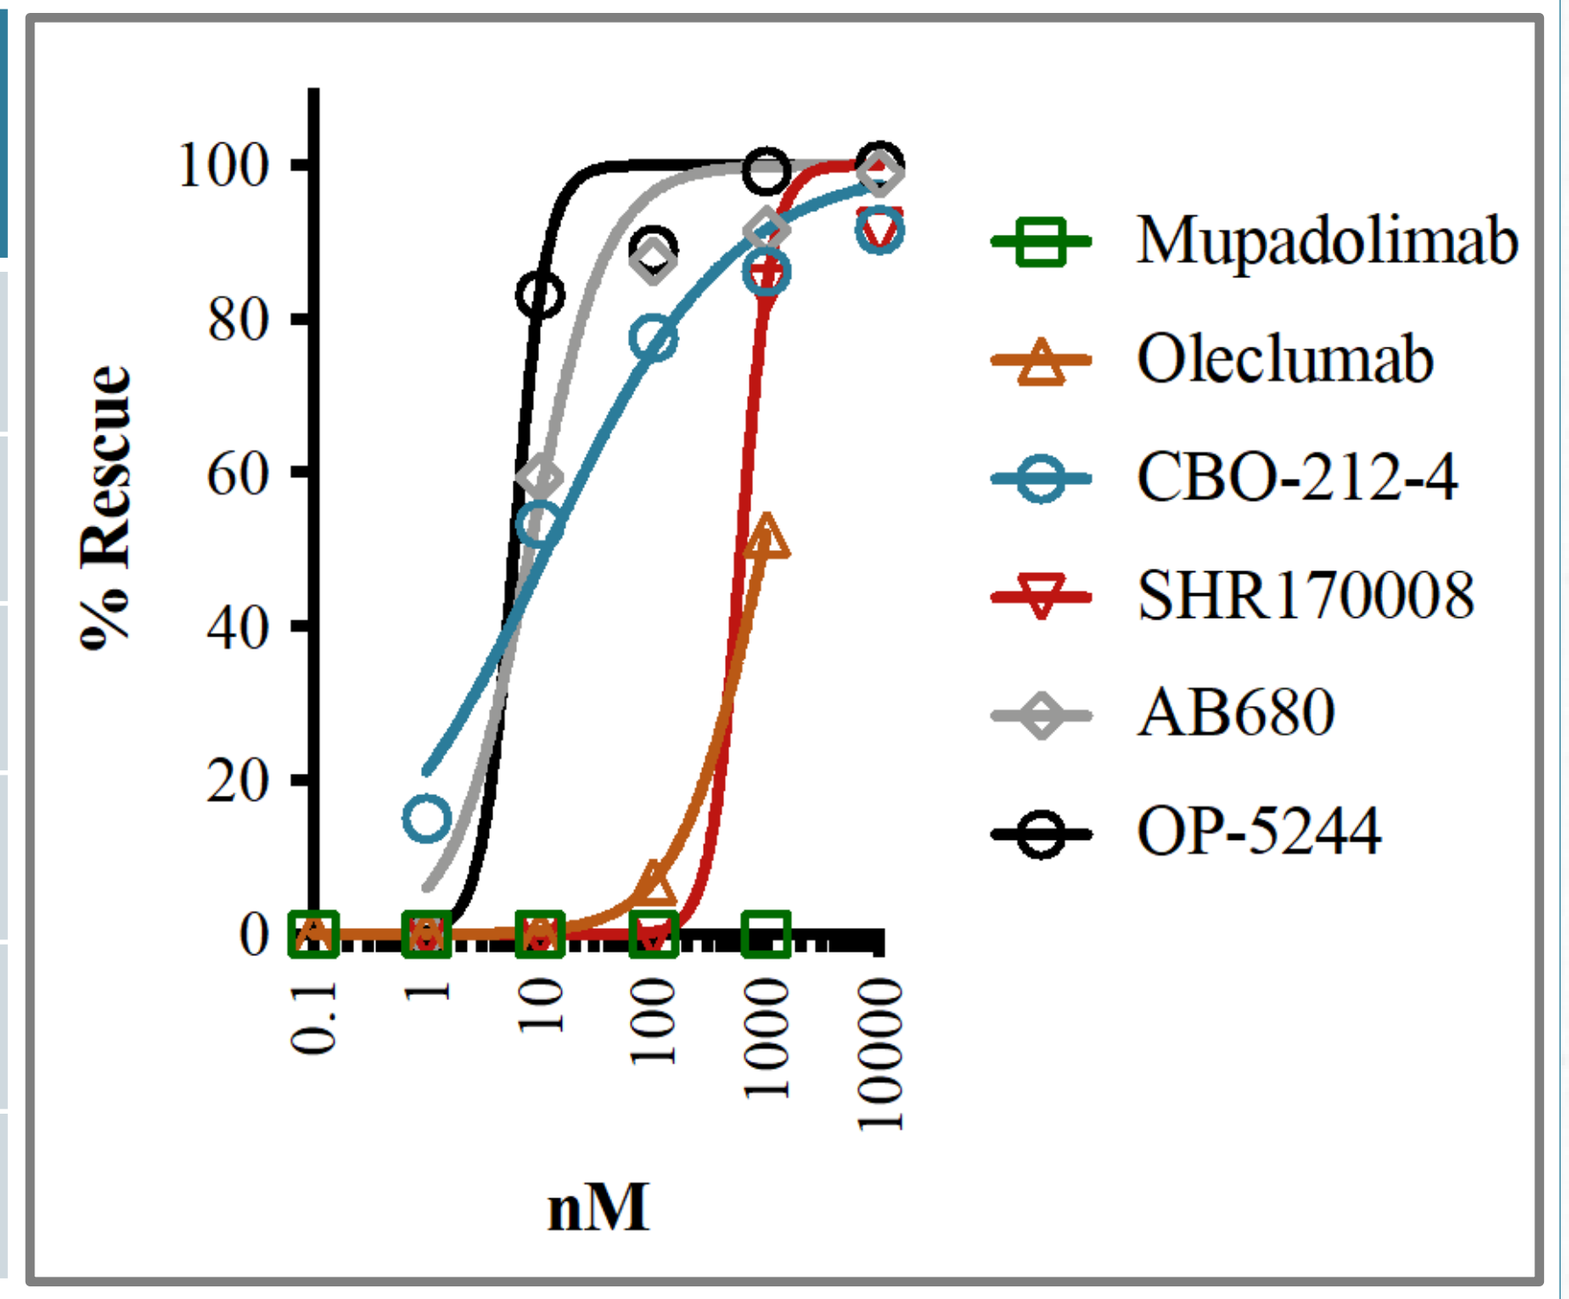
% Rescue
nM
Mupadolimab
Oleclumab
CBO-212-4
SHR170008
AB680
OP-5244

In a functional assay the ability of test articles to rescue human PBMCs suppressed with AMP was determined using the CD25 T cell activation marker, and granzyme B required for cancer cell killing. CBO-212 demonstrated increased potency compared to both mAbs and similar or improved activity to the small molecule comparators (**Figure 3**).

RESULTS (cont.)

Figure 4. *CD73 internalization assay with CBO-212 vs comparators (EC_{50} in nM)*

Test article	EC_{50} [nM]
CBO-212	0.11
AB680	>100
OP-5244	>100
SHR170008	>100
Oleclumab	0.02
Mupadolimab	0.06

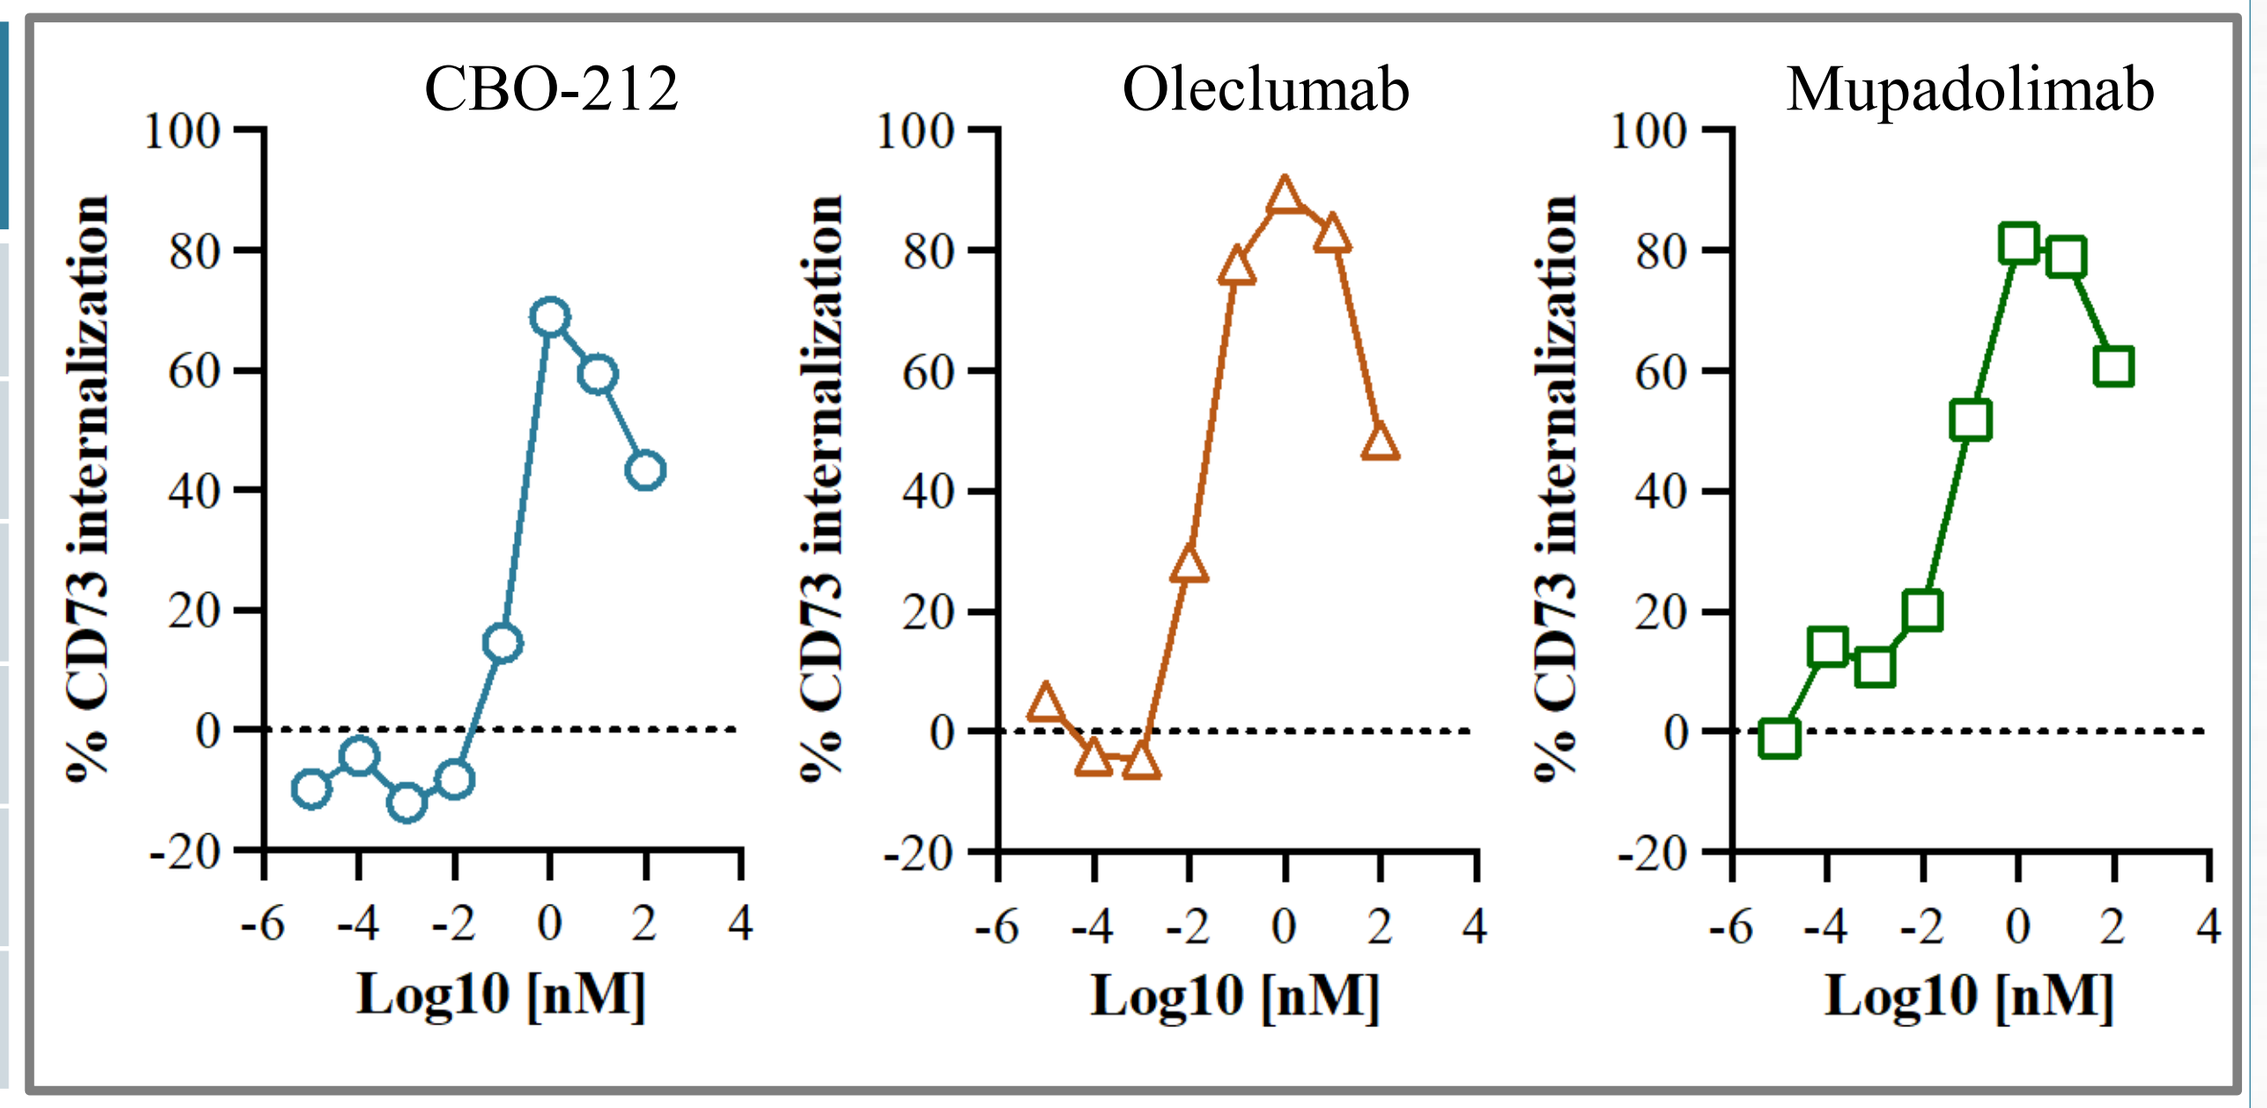
CBO-212
Oleclumab
Mupadolimab
% CD73 internalization
Log10 [nM]

CD73 internalization is an additional mechanism to potentially reduce adenosine levels in the TME that is only achievable via receptor cross-linking, which is not possible with small molecule inhibitors. Because DFCs present multiple small molecule inhibitors, we hypothesized CBO-212 could promote receptor cross-linking to stimulate internalization. CBO-212 induced receptor internalization in a manner similar to the mAb comparators (**Figure 4**). CBO-212 showed a dose-dependent increase in CD73 internalization followed by reduction at high concentrations.

The PK and stability of CBO-212 were investigated in BALB/c mice administered a single IP injection at 3 mg/kg. Over one week CBO-212 levels remained high, consistent with a long circulating half-life in mice (**Figure 5**). To determine the stability of CBO-212 *in vivo*, dual-capture sandwich ELISA methods were used. Briefly, side-by-side assays were conducted using either CD73 or Fc as the primary capture method, followed by Fc detection. Comparing the AUC ratios from both capture methods allows for evaluation of retention of functional conjugated small molecule inhibitors on the conjugate. The nearly superimposable PK curves generated using the two capture methods demonstrate that circulating CBO-212 remained intact and unchanged for the study duration.

Figure 5. *PK and stability of CBO-212 in BALB/c mice*

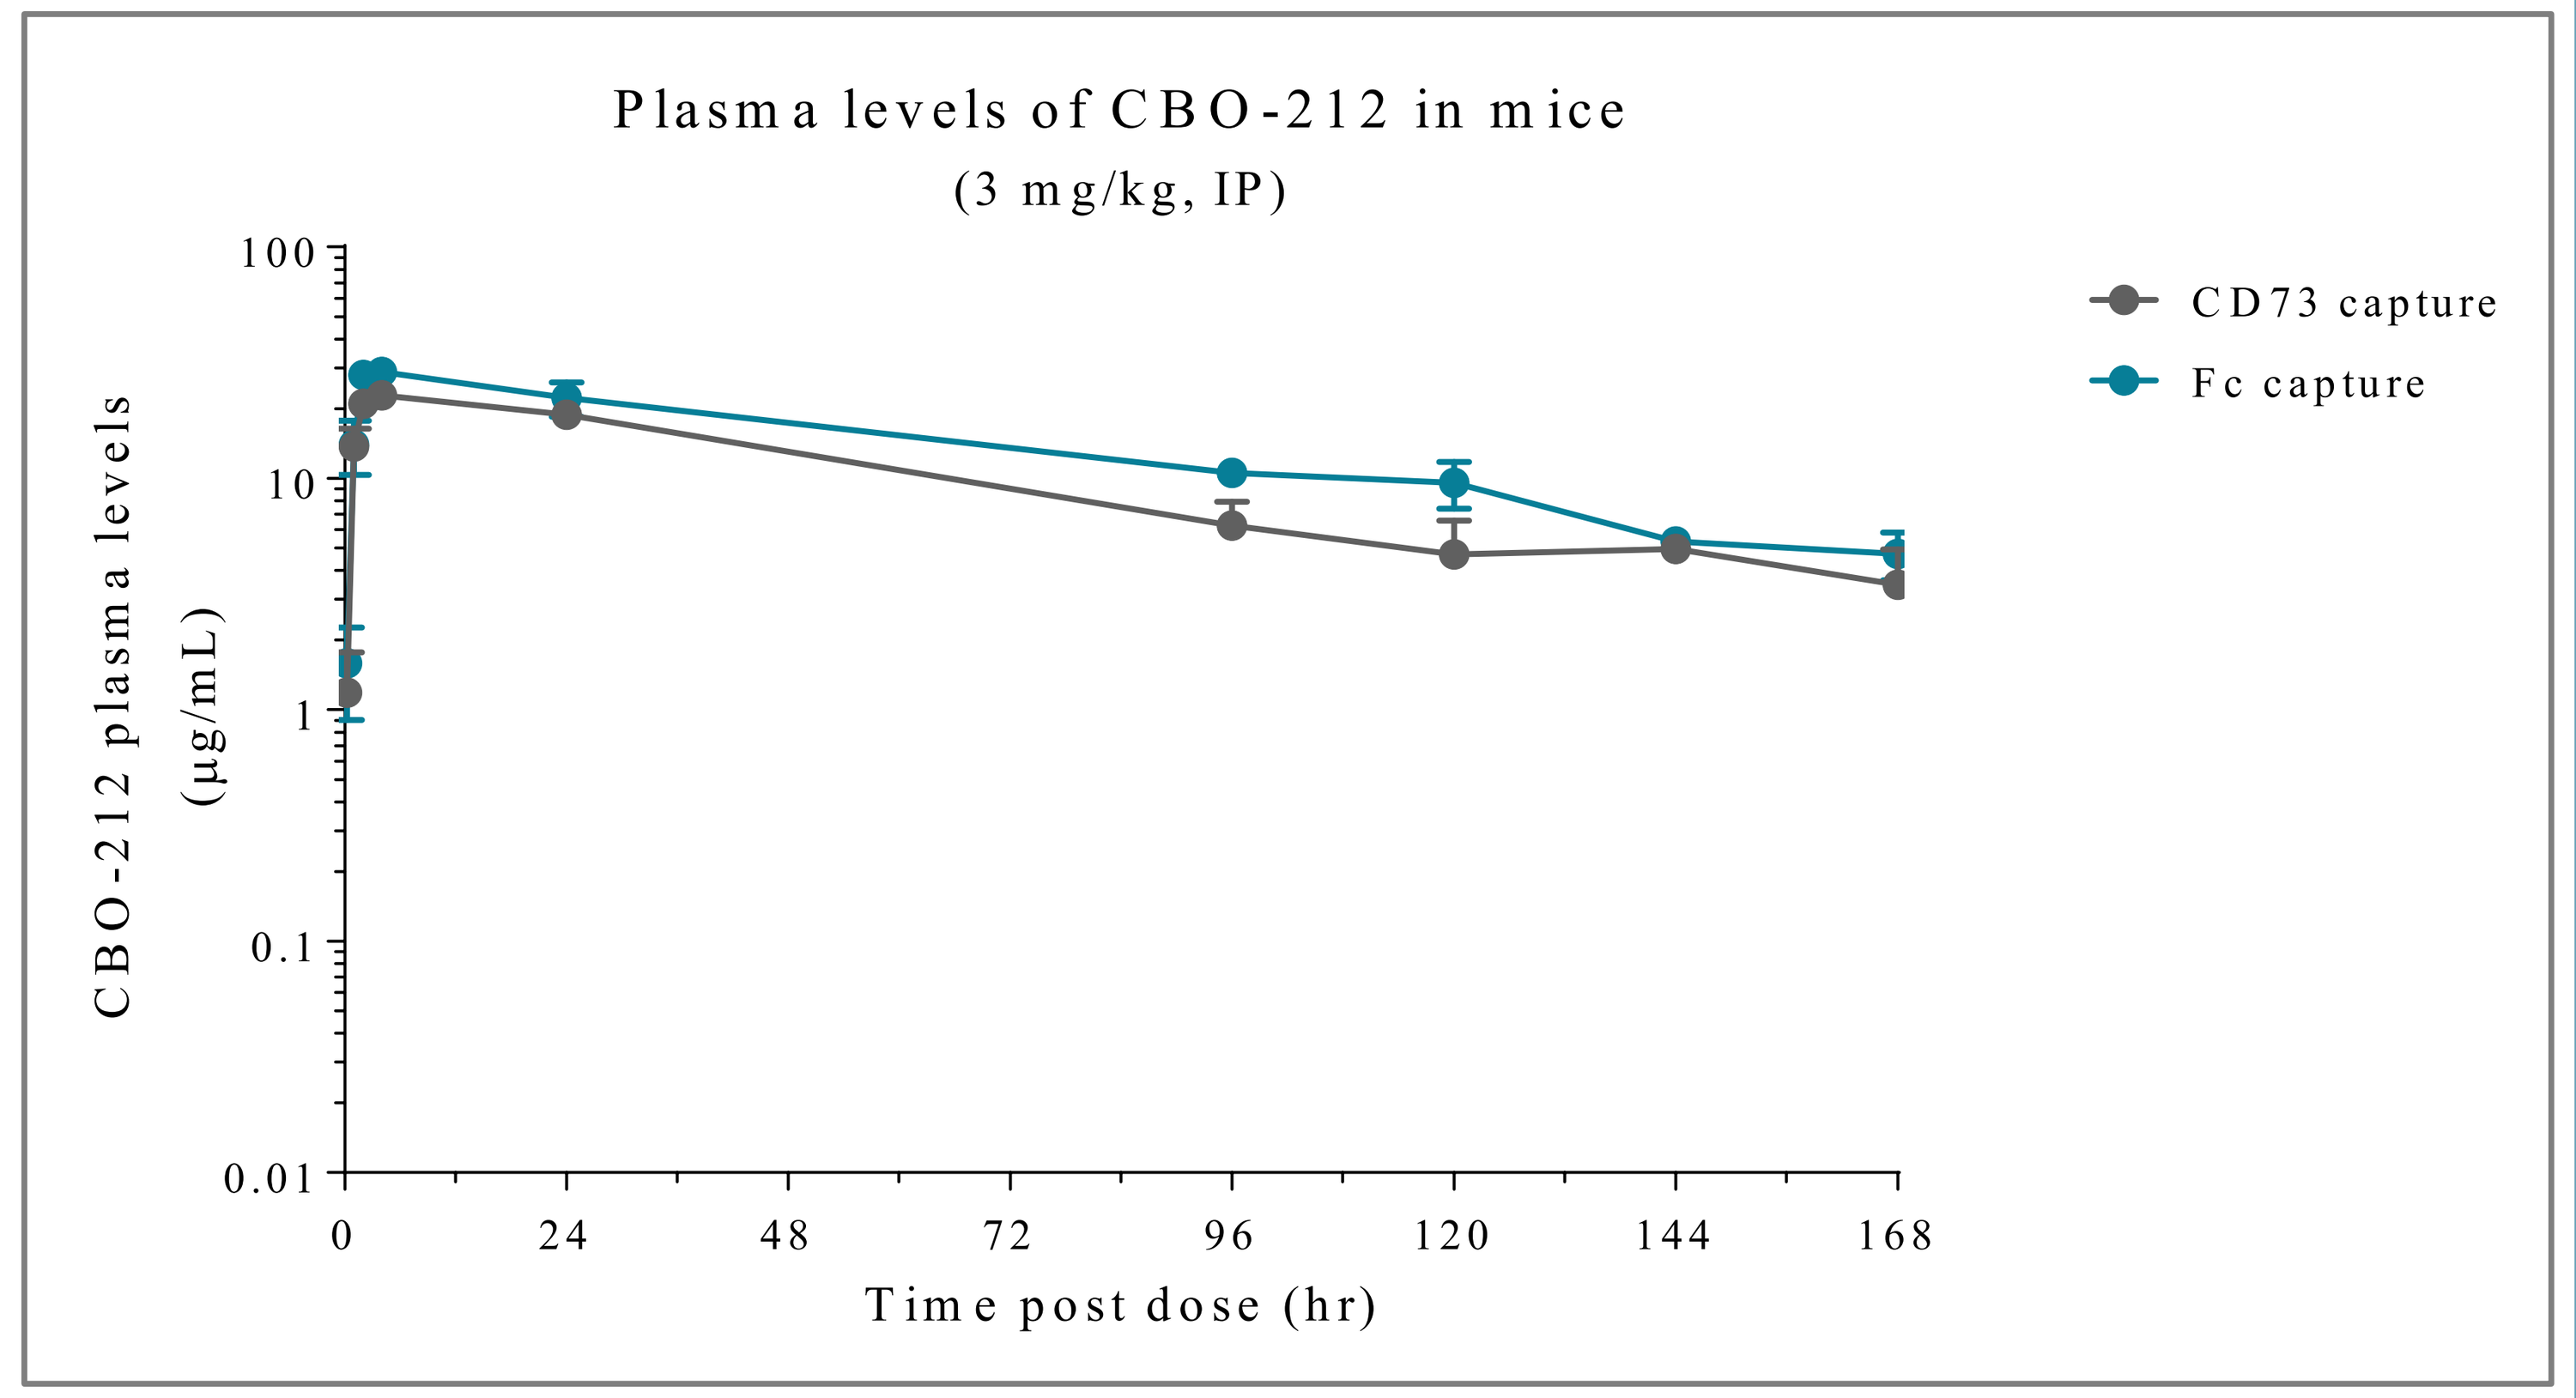
Plasma levels of CBO-212 in mice
(3 mg/kg, IP)
CBO-212 plasma levels
(µg/mL)
Time post dose (hr)
CD73 capture
Fc capture

RESULTS (cont.)

Figure 6. *Efficacy of a single CBO-212 dose in a syngeneic mouse model (CT26).*

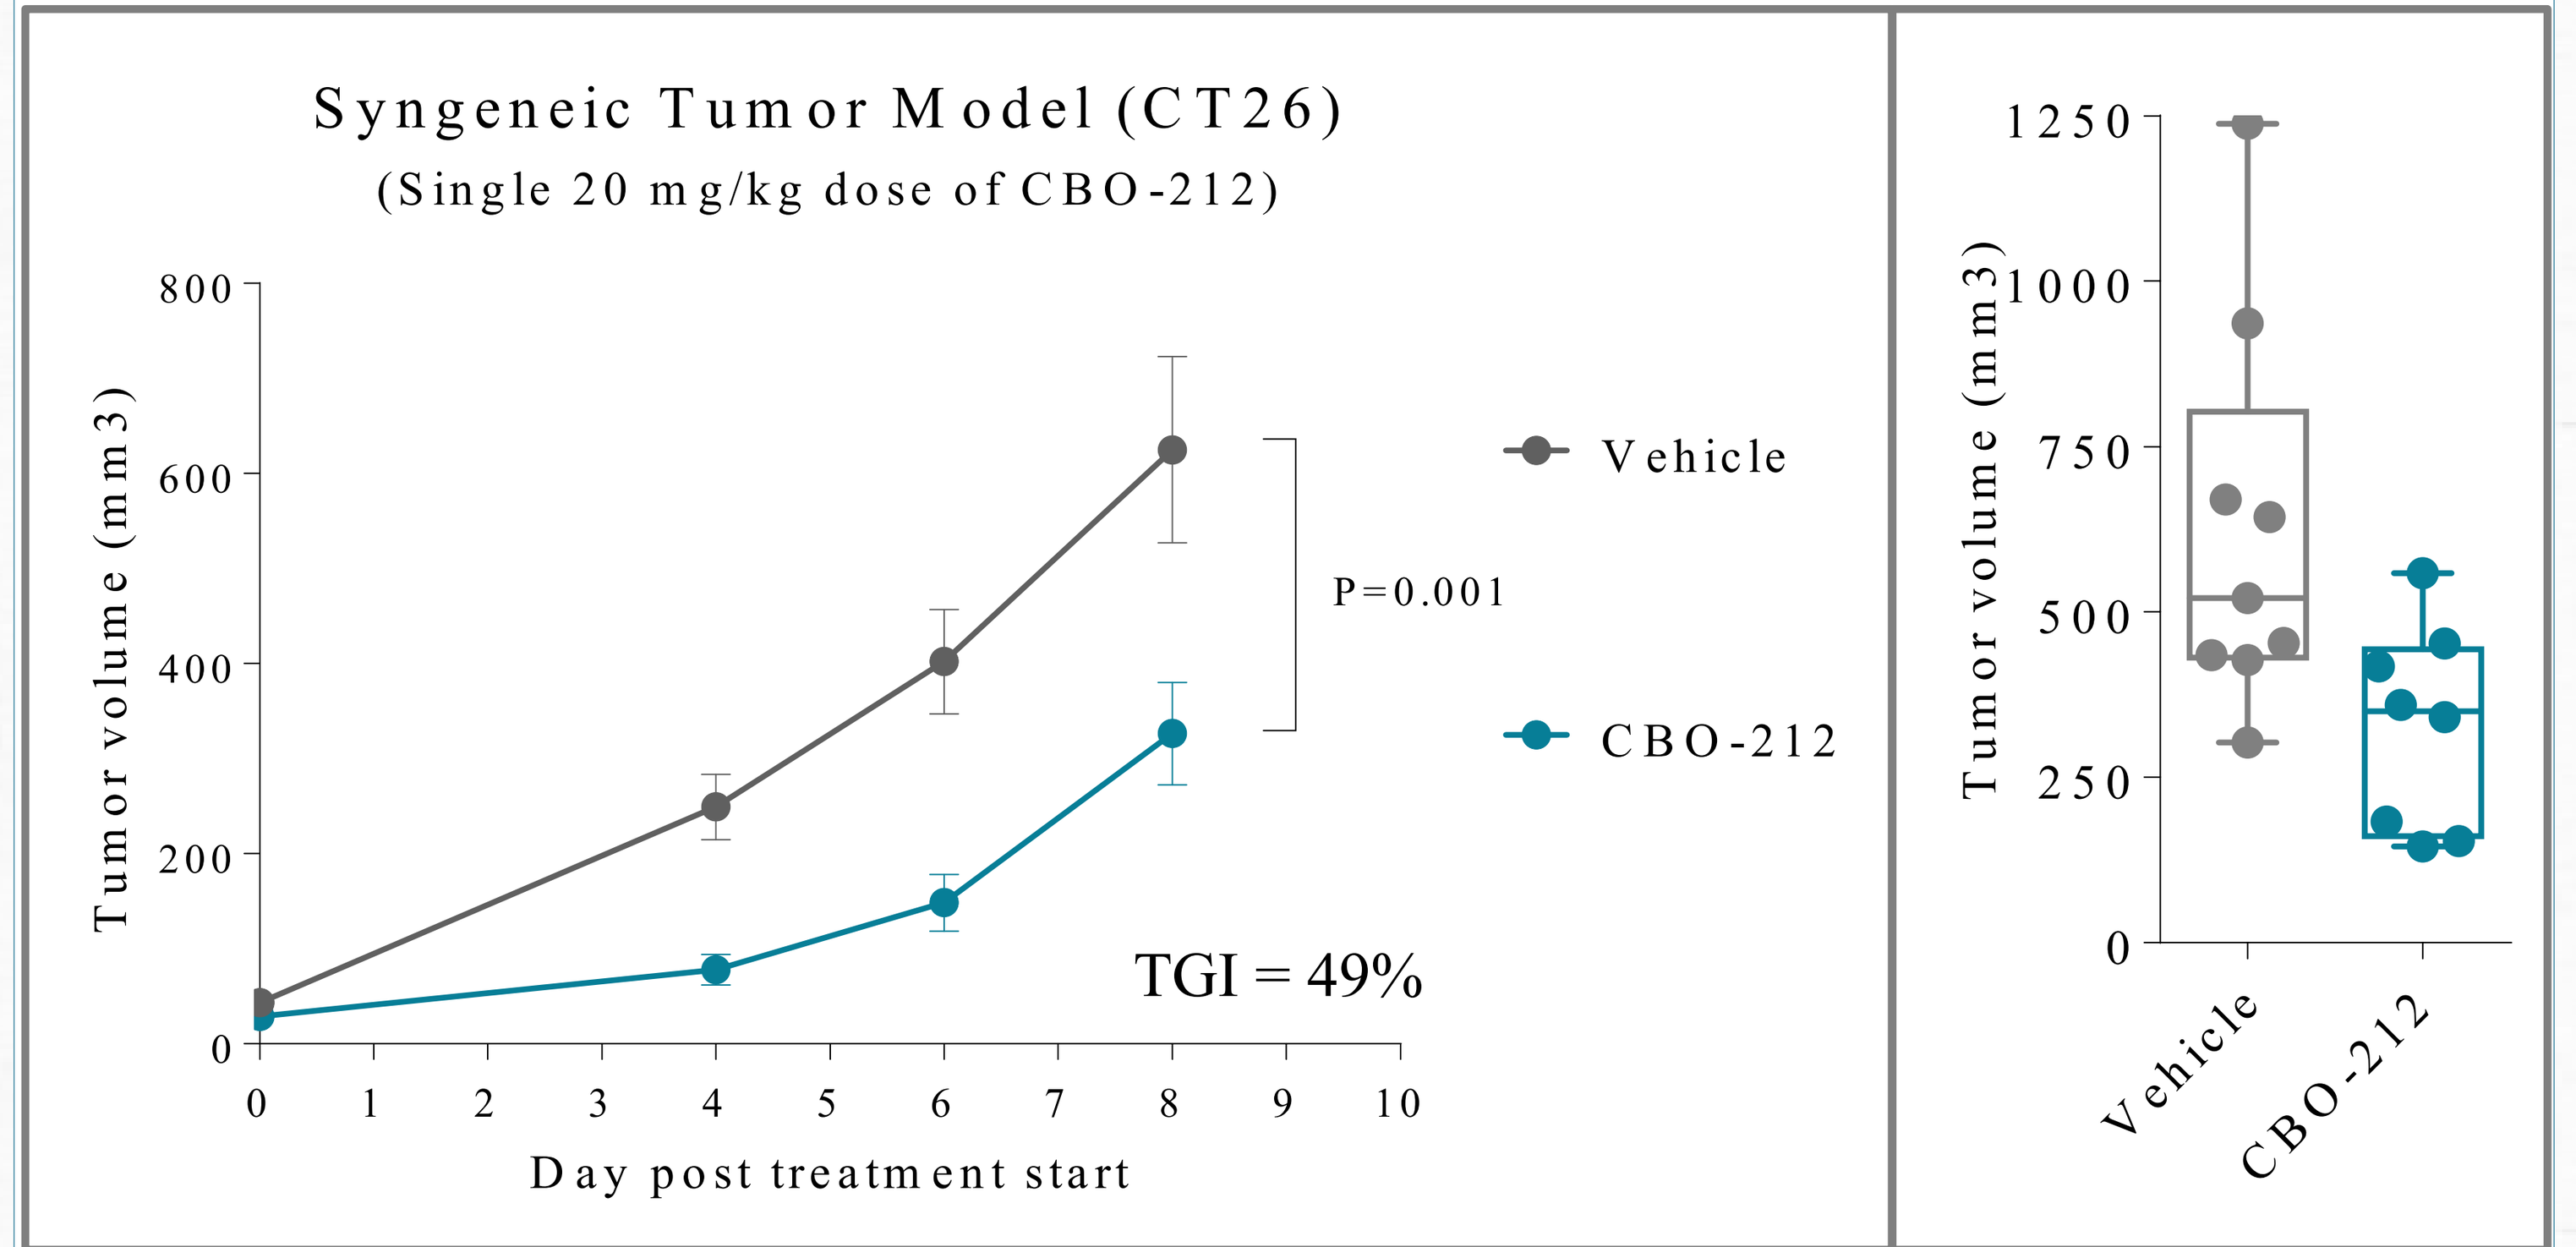
Syngeneic Tumor Model (CT26)
(Single 20 mg/kg dose of CBO-212)
Tumor volume (mm3)
Day post treatment start
Vehicle
CBO-212
P=0.001
TGI = 49%

CBO-212 demonstrated efficacy in a syngeneic mouse tumor model using the colon cancer cell line CT26. In this study mice with established tumors (25 – 50 mm^3) were treated with a single 20 mg/kg IP dose of CBO-212. This resulted in significant reduction in tumor growth over time relative to vehicle treated animals (**Figure 6**). Compared to vehicle this resulted in a 49% TGI. Based on these results additional studies are underway to evaluate the efficacy of CBO-212 in mono- and combination therapy settings against multiple murine cancer cell-lines.

CONCLUSIONS

CBO-212 is an AMP competitive, catalytic site inhibitor that demonstrated high potency in CD73 enzyme inhibition and functional PBMC rescue assays. The *in vitro* potency and favorable PK of CBO-212 translated to significant TGI in a syngeneic tumor model with a single dose. Based in part on these results, CBO-212 is being advanced as a clinical development candidate for the treatment of solid cancers.

Table 2. *Potential for differentiation of CBO-212 vs other therapeutic modalities*

Activity	Small molecule	mAb	DFC
Soluble CD73 inhibition	+++	–/+	+++
Cell-anchored CD73 inhibition	+++	–/++	+++
Receptor internalization	–	–/+++	+++
Half-Life	+	+++	+++
Tissue/tumor penetration	+++	+	++
Potential safety profile*	++	+++	+++
Ability to combine MOAs	-	+	+++

*mAbs and DFCs do not enter the intracellular space, reducing potential for off-target toxicities

DISCLOSURES

All authors are shareholders and/or employees of Cidara Therapeutics.