2116

EFFICACY OF CD388, A NOVEL DRUG FC-CONJUGATE (DFC), IS DRIVEN BY THE SMALL MOLECULE NEURAMINIDASE INHIBITOR (NAI)

Simon Döhrmann*, James Levin, Elizabeth Abelovski, Amanda Almaguer, Rajvir Grewal, Karin Amundson, Joanne Fortier, Thanh Lam, Thomas P. Brady, Allen Borchardt, Jason N. Cole, Leslie W. Tari

Cidara Therapeutics, 6310 Nancy Ridge Drive, San Diego, CA, 92121 USA

*corresponding author
sdoehrmann@cidara.com

BACKGROUND

Influenza prevention remains a significant public health concern that is still not adequately addressed by vaccines or current therapeutic options. Cidara has developed CD388, a multivalent conjugate of a dimeric NAI with a proprietary hIgG1 Fc domain engineered for extended half-life. CD388 is in clinical development (NCT05285137 and NCT05523089) for the prevention of seasonal influenza A and B. Herein, we analyze the contribution of Fc-mediated immune effector function to CD388 efficacy in lethal mouse models.

METHODS

CD388, Fc engineered to extend half-life, and closely related Fc modified analogues (immune-active 'IA' or immune-silent 'IS') were generated. CD388-IA engages Fc gamma receptors (FcgRs) whereas IS fails to engage FcgRs required to trigger Fc-mediated immune effector functions (e.g. antibody-dependent cellular cytotoxicity). Efficacy studies were conducted in BALB/c WT or *Fcer1g* KO mice. The *Fcer1g* KO mice are deficient in activating FcgRs thereby preventing contribution of Fc-mediated immunity to efficacy. After lethal challenge with mouse-adapted influenza A/Puerto Rico/8/1934 (H1N1) virus, CD388 or analogues were administered SC or IM two hours post-infection. Animals were monitored daily for 14 days for survival (<20% BW loss). For cytokine and viral load quantification, lungs were harvested after 4 days and viral load determined by plaque assay.

RESULTS

CD388-IA and -IS at comparable drug to antibody ratios (DARs) of 4.5±0.5, same DAR range as used in the clinics, demonstrated comparable protection (**Figure 1A, D**) and comparable dose-dependent viral burden and pro-inflammatory cytokine reduction for MCP-1 (**Table 1**) and IL-6, MIP-1α (*data not shown*) in a lethal mouse model. Additionally, the CD388-IA analogue at DAR 4.5±0.5 was protective in *Fcer1g* KO mice at the same doses required for protection in WT mice. However, a CD388-IA analogue at low DAR of 1, that had reduced antiviral activity (*data not shown*), demonstrated overall reduced efficacy as compared to DAR 4.5±0.5, but improved efficacy with minimal protective dose of 0.3 mg/kg in WT mice as compared to 1 mg/kg in *Fcer1g* KO mice (**Figure 1B-C, E-F**) suggesting Fc-mediated contribution at low DAR.

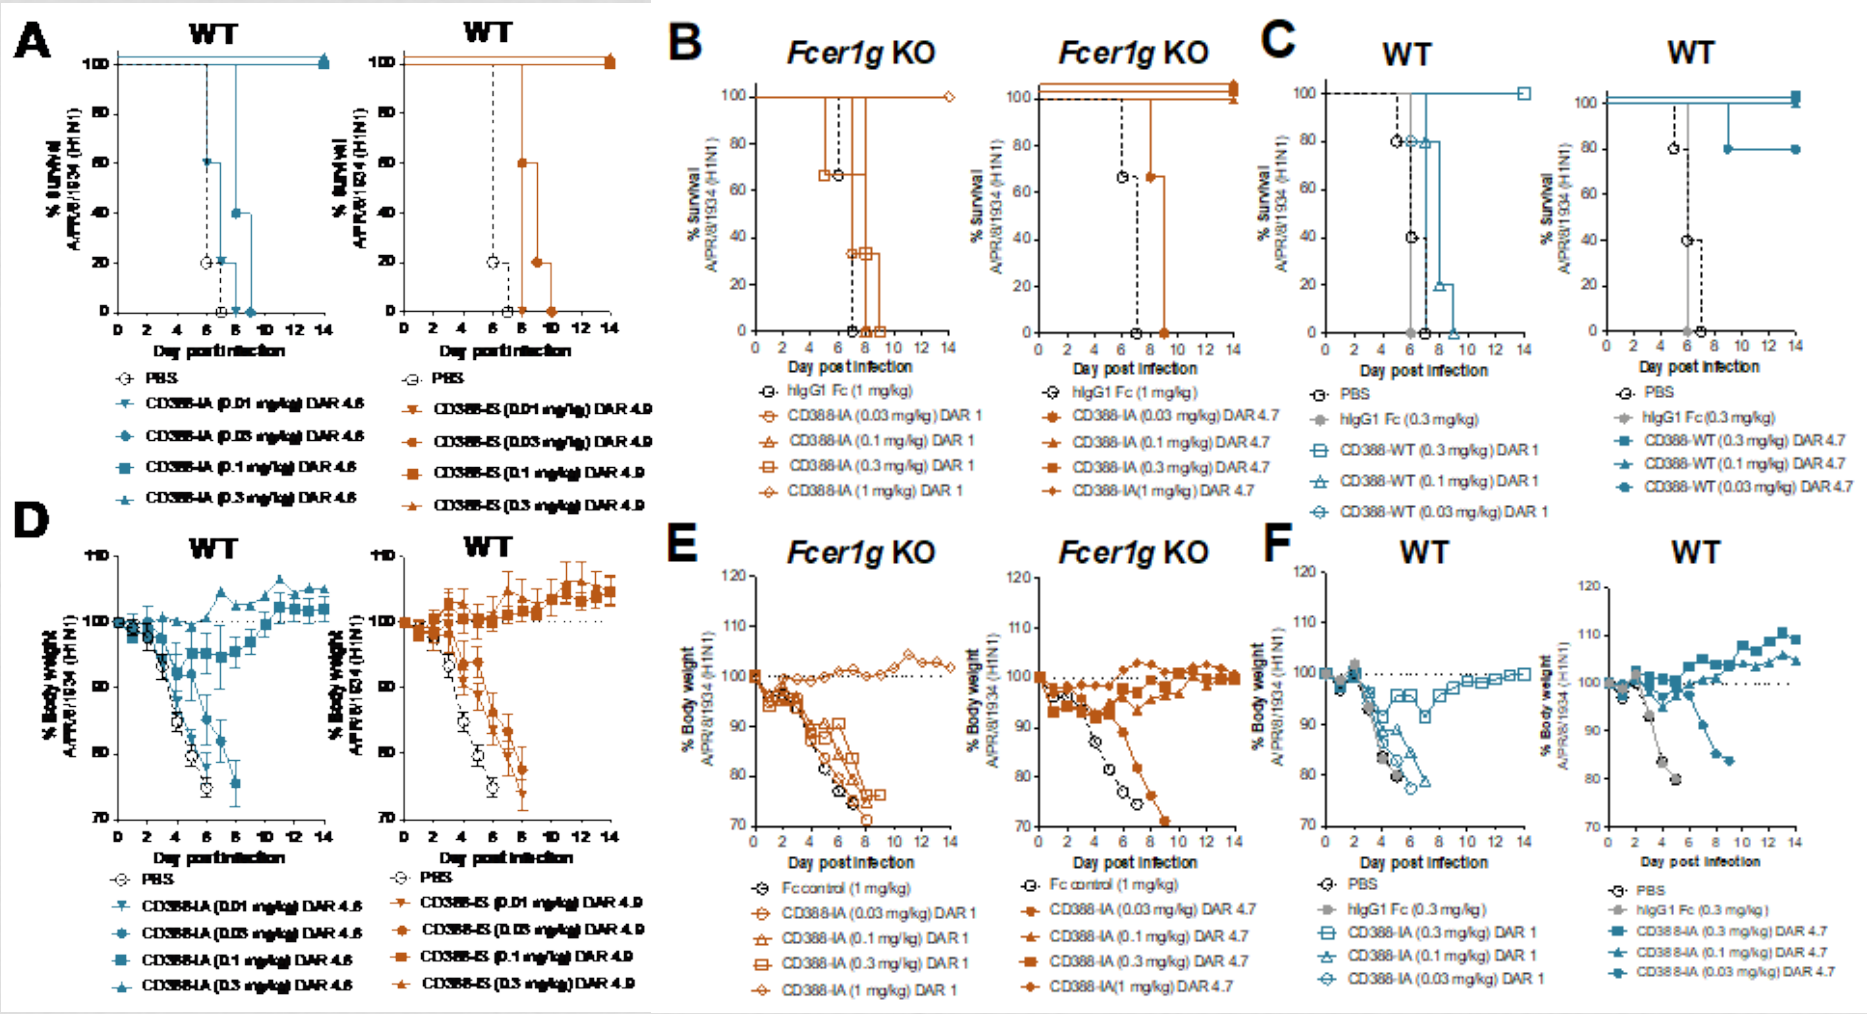
A
WT
WT
Day post infection
PBS
CD388-IA (0.01 mg/kg) DAR 4.6
CD388-IA (0.03 mg/kg) DAR 4.6
CD388-IA (0.1 mg/kg) DAR 4.6
CD388-IA (0.3 mg/kg) DAR 4.6
CD388-IS (0.01 mg/kg) DAR 4.9
CD388-IS (0.03 mg/kg) DAR 4.9
CD388-IS (0.1 mg/kg) DAR 4.9
CD388-IS (0.3 mg/kg) DAR 4.9
B
Fcer1g KO
Fcer1g KO
% Survival A/PR/8/1934 (H1N1)
Day post infection
hIgG1 Fc (1 mg/kg)
CD388-IA (0.03 mg/kg) DAR 1
CD388-IA (0.1 mg/kg) DAR 1
CD388-IA (0.3 mg/kg) DAR 1
CD388-IA (1 mg/kg) DAR 1
CD388-IA (0.03 mg/kg) DAR 4.7
CD388-IA (0.1 mg/kg) DAR 4.7
CD388-IA (0.3 mg/kg) DAR 4.7
CD388-IA(1 mg/kg) DAR 4.7
C
WT
WT
PBS
hIgG1 Fc (0.3 mg/kg)
CD388-WT (0.3 mg/kg) DAR 1
CD388-WT (0.1 mg/kg) DAR 1
CD388-WT (0.03 mg/kg) DAR 1
CD388-WT (0.3 mg/kg) DAR 4.7
CD388-WT (0.1 mg/kg) DAR 4.7
CD388-WT (0.03 mg/kg) DAR 4.7
D
WT
WT
E
Fcer1g KO
Fcer1g KO
% Body weight A/PR/8/1934 (H1N1)
Fc control (1 mg/kg)
F
WT
WT
CD388-IA (0.3 mg/kg) DAR 1
CD388-IA (0.1 mg/kg) DAR 1
CD388-IA (0.03 mg/kg) DAR 1
CD388-IA (0.3 mg/kg) DAR 4.7
CD388-IA (0.1 mg/kg) DAR 4.7
CD388-IA (0.03 mg/kg) DAR 4.7

Figure 1. Efficacy of (A) CD388-IA (DAR 4.6) or CD388-IS (DAR 4.9) in WT mice. Efficacy of CD388-IA at DAR 1 or DAR 4.7 in (B) *Fcer1g* KO, or (C) WT mice against A/PR/8/1934 (H1N1) in a lethal mouse model showing survival (A-C) and corresponding body weight (D-F).

RESULTS

Table 1. Viral load reduction and MCP-1 analysis (fold-change vs uninfected control) on day 4.

Treatment [mg/kg]	Log Viral Load Reduction	MCP-1
PBS	0.00	30.0
Fc only [3]	-0.11	31.9
CD388-IA [0.03]	0.07	14.8
CD388-IA [0.1]	0.54	10.2
CD388-IA [0.3]	1.92	3.2
CD388-IA [1]	3.34	1.2
CD388-IA [3]	4.16	1.2
CD388-IS [0.03]	0.38	19.4
CD388-IS [0.1]	0.67	11.1
CD388-IS [0.3]	2.21	10.7
CD388-IS [1]	2.68	4.7
CD388-IS [3]	3.57	4.5
Uninfected	n.d.	1.0

CONCLUSION

These data demonstrate that the efficacy of CD388 at DAR 4.5±0.5 is driven predominantly by the intrinsic antiviral activity of the small molecule NAIs and is largely independent of contributions from Fc-mediated effector function. These data suggest that CD388 has the potential to demonstrate equivalent activity in healthy and immune compromised individuals

DISCLOSURES

All authors are shareholders and employees of Cidara Therapeutics.