2514

SINGLE ASCENDING DOSE SAFETY, TOLERABILITY, AND PHARMACOKINETICS OF SUBCUTANEOUS AND INTRAMUSCULAR CD388, A NOVEL LONG-ACTING DRUG-FC CONJUGATE FOR UNIVERSAL PREVENTION OF SEASONAL AND PANDEMIC INFLUENZA

Shawn Flanagan
sflanagan@cidara.com
858.249.7686

Shawn Flanagan, [1] Voon Ong, Tristan Baguet,[2] Roxana Rojas,[2] Sy-Shi Wang,[2] Ozlem Equils[1]

[1]Cidara Therapeutics Inc. [2]Janssen R&D

BACKGROUND

CD388 is a novel, long-acting, antiviral drug-Fc conjugate (DFC) designed to deliver universal prevention of seasonal and pandemic influenza. Nonclinical studies have shown CD388 to be effective prophylactically against seasonal and pandemic influenza A and B strains in lethal and nonlethal mouse models of infection.

METHODS

The design of the ongoing blinded CD388 First in Human study (NCT05285137), which is being conducted in healthy subjects is shown in Figure 1. CD388 for injection (100 mg/mL) was administered by intramuscular (IM) or subcutaneous (SQ) route. Safety and tolerability data were obtained in all subjects. Single dose CD388 pharmacokinetics were determined for subjects in the 50, 150, and 450 mg cohorts of each dosing route at protocol specified nominal time points; alias subject numbers were used to maintain blind. 900 mg SQ cohort still ongoing. Plasma CD388 concentrations were determined for subjects randomized to receive CD388 using a validated hybrid immunoassay LC-MS method with lower limit of quantitation (LLOQ) of 0.1 µg/mL. Noncompartmental pharmacokinetics were determined using Phoenix WinNonlin (Certara NC, USA).

Figure 1: Design of Ongoing CD388 First in Human Study

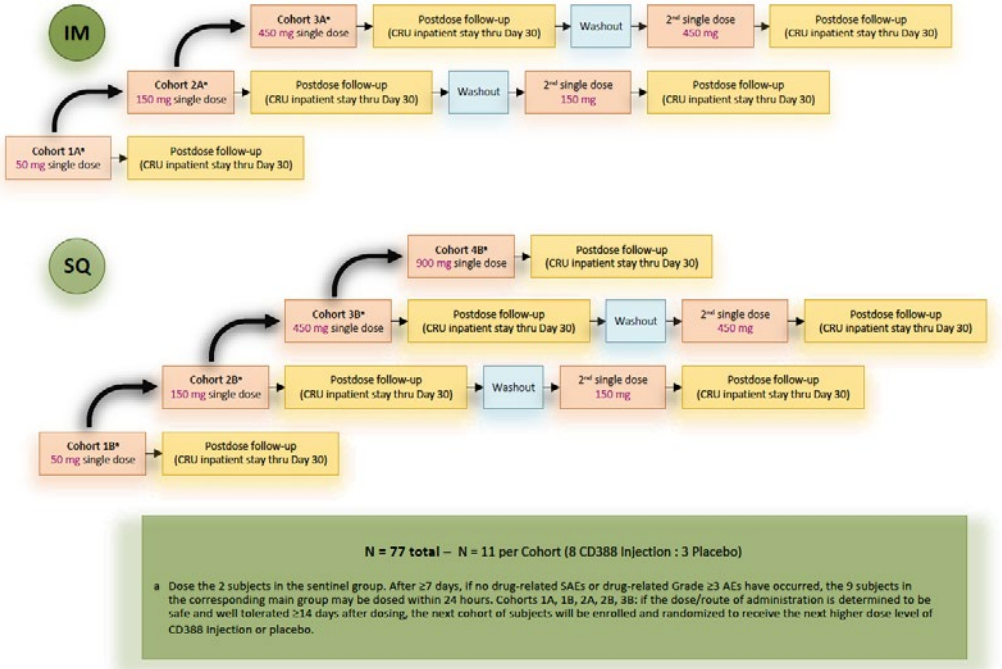
IM
Cohort 3A* 450 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
Washout
2nd single dose 450 mg
Postdose follow-up (CRU inpatient stay thru Day 30)
Cohort 2A* 150 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
Washout
2nd single dose 150 mg
Postdose follow-up (CRU inpatient stay thru Day 30)
Cohort 1A* 50 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
SQ
Cohort 4B* 900 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
Cohort 3B* 450 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
Washout
2nd single dose 450 mg
Postdose follow-up (CRU inpatient stay thru Day 30)
Cohort 2B* 150 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
Washout
2nd single dose 150 mg
Postdose follow-up (CRU inpatient stay thru Day 30)
Cohort 1B* 50 mg single dose
Postdose follow-up (CRU inpatient stay thru Day 30)
N = 77 total – N = 11 per Cohort (8 CD388 Injection : 3 Placebo)
a Dose the 2 subjects in the sentinel group. After ≥7 days, if no drug-related SAEs or drug-related Grade ≥3 AEs have occurred, the 9 subjects in the corresponding main group may be dosed within 24 hours. Cohorts 1A, 1B, 2A, 2B, 3B: if the dose/route of administration is determined to be safe and well tolerated ≥14 days after dosing, the next cohort of subjects will be enrolled and randomized to receive the next higher dose level of CD388 Injection or placebo.

RESULTS

Safety and Tolerability

- Overall profile: well-tolerated up to 900 mg single dose; study in progress with limited follow up at highest doses
- No treatment-emergent SAEs and no withdrawals due to safety findings
- Most TEAEs were Grade 1 (90%), few Grade 2, all resolved within the exposure time-frame
- Incidence of TEAE not dose-dependent
- No hypersensitivity reactions were observed
- Few injection site events (pain, IM route mainly), Grade 1, all resolved spontaneously
- No clinically relevant ECG, vital signs or physical exam abnormalities

Pharmacokinetics

- IM or SQ CD388 was well absorbed
- Time of maximum plasma concentrations (T_{max}) appeared from 2 to 13 days post dose, and quantifiable concentrations (>LLOQ) lasted for months (Figure 2)
- CD388 AUC increased approximately dose proportional for both routes (Figure 3)
- Mean apparent half-life of elimination was ~ 6 to 7 weeks (Table 1)

Figure 2: Single Dose Mean Plasma CD388 Concentrations versus Time for 50, 150, or 450 mg CD388 Administered by Intramuscular or Subcutaneous Injection (Semi-logarithmic Scale)

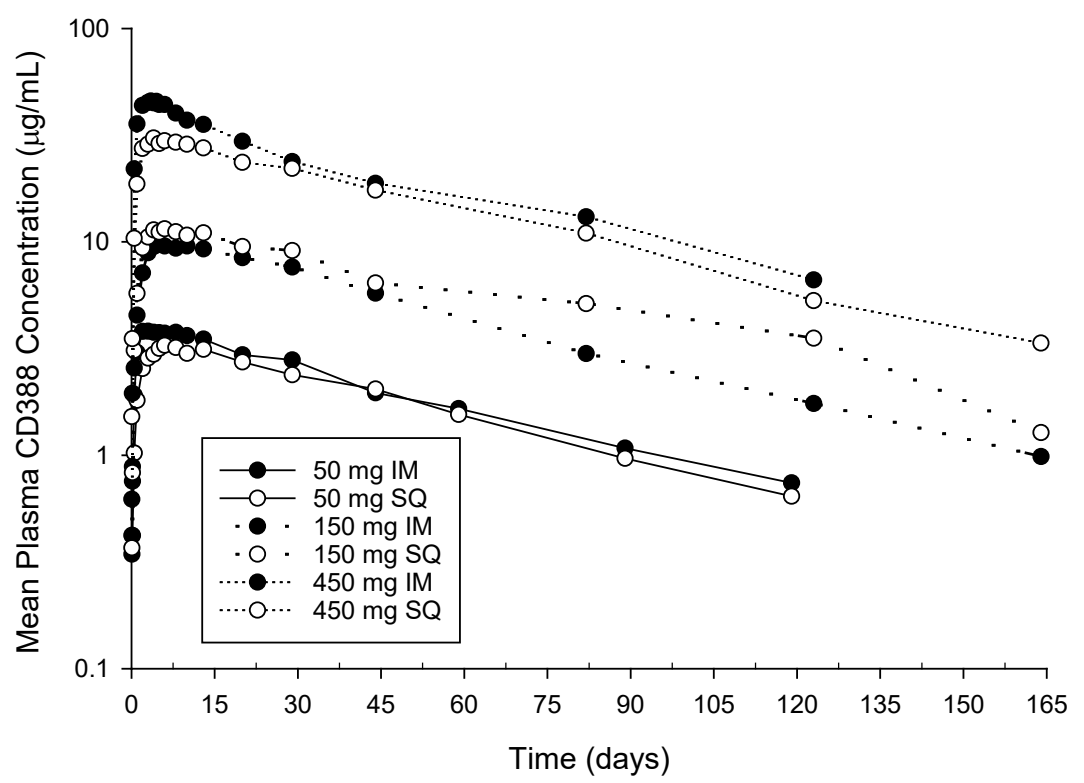
Mean Plasma CD388 Concentration (µg/mL)
100
10
1
0.1
0
15
30
45
60
75
90
105
120
135
150
165
Time (days)
50 mg IM
50 mg SQ
150 mg IM
150 mg SQ
450 mg IM
450 mg SQ

Table 1: Plasma CD388 PK Parameter Estimates

Group	T_{max} (day)	C_{max} (µg/mL)	$AUC_{0-\infty}$ (µg•day/mL)	$t_{1/2}$ (day)
IM 50 mg	4.0, 2-10	4.1 (18.1%)	280 (15.9%)	50.4 (18.4%)
SQ 50 mg	9.5, 3-13	3.6 (26.4%)	246 (25.6%)	46.3 (18.1%)
IM 150 mg	5.5, 3-13	10.4 (23.4%)	738 (18%)	47.5 (23.2%)
SQ 150 mg	5.5, 2-13	12 (27.8%)	879 (18.6%)	53.3 (19%)
IM 450 mg	3.3, 1-6	48.6 (24.2%)	2579 (22.5%)	43.8 (25.2%)
SQ 450 mg	4.0, 2-10	32.7 (28.7%)	2254 (31.7%)	45.0 (23.2%)

Data are presented as mean (standard deviation) for N= 8 subjects per group, except T_{max}= Median, Range.

Figure 3: Mean CD388 AUC Versus Intramuscular or Subcutaneous Dose

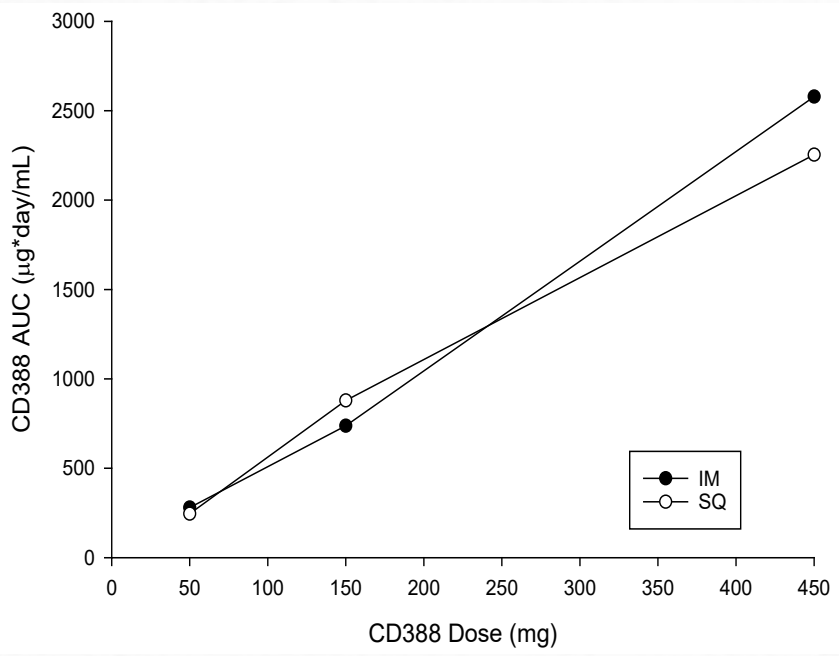
CD388 AUC (µg*day/mL)
3000
2500
2000
1500
1000
500
0
0
50
100
150
200
250
300
350
400
450
CD388 Dose (mg)
IM
SQ

CONCLUSIONS

- CD388 was generally well tolerated with no significant safety concerns in this ongoing study at single doses of up to 900 mg
- Plasma concentrations and pharmacokinetics of CD388 were similar following IM or SQ routes of administration
- Sustained plasma CD388 concentrations due to its long half-life indicate potential for season-long protection with a single dose

DISCLOSURES / ACKNOWLEDGMENTS

This study was funded by Janssen R&D.